Encaleret normalizes mineral homeostasis and is associated with an increase in eGFR in patients with autosomal dominant hypocalcemia Type 1 (ADH1): 24-week phase 3 CALIBRATE results

SA-PO0551

John A. Sayer[1,2], Andrea Palermo[3], Rachel Gafni[4], Steven Ing[5], Aliya A. Khan[6], Michael A. Levine[7], Nobuaki Ito[8], Erik Imel[9], Peter Kamenicky[10], Lars Rejnmark[11], Mark L. Warren[12], Robert A. Wermers[13], Filomena Cetani[14], Abhishek Kansara[15], Sandrine Lemoine[16,17], Vit Zikan[18], Roderick J. Clifton-Bligh[19,20], Nina S. Ma[21,22], Mishaela Rubin[23], Evert F. van Velsen[24], Jessica Chen[25], Ajay Singh[25], Mary S. Roberts[25], Scott H. Adler[25], Michael Mannstadt[26,27]

[1]Newcastle University, Newcastle upon Tyne, England, United Kingdom; [2]Newcastle Upon Tyne NHS Foundation Trust, Newcastle upon Tyne, United Kingdom; [3]Policlinico Universitario Campus Bio-Medico, Rome, Italy; [4]National Institutes of Health, Bethesda, MD, United States; [5]The Ohio State University Wexner Medical Center, Columbus, OH, United States; [6]McMaster University, Hamilton, ON, Canada; [7]Children's Hospital of Philadelphia, Philadelphia, PA, United States; [8]The University of Tokyo, Graduate School of Medicine and Faculty of Medicine, Tokyo, Japan; [9]Indiana University School of Medicine, Indianapolis, IN, United States; [10]Université Paris-Saclay, AP-HP, Hôpital Bicêtre, Le Kremlin-Bicêtre, France; [11]Aarhus Universitetshospital, Aarhus, Denmark; [12]Physicians East PA, Greenville, NC, United States; [13]Mayo Clinic, Department of Medicine, Rochester, MN, United States; [14]University of Pisa, Department of Clinical and Experimental Medicine, Pisa, Italy; [15]Houston Methodist, Department of Medicine, Houston, TX, United States; [16]Edouard Herriot Hospital, Department of Nephrology and Physiology, Lyon, France; [17]Université de Lyon, Lyon, France; [18]Charles University, General University Hospital and Faculty of Medicine 1, Prague, Czechia; [19]Royal North Shore Hospital, Sydney, NSW, Australia; [20]University of Sydney, Faculty of Medicine and Health, Sydney, NSW, Australia; [21]University of Colorado Anschutz Medical Campus School of Medicine, Aurora, CO, United States; [22]Children's Hospital Colorado, Pediatric Endocrinology, Aurora, CO, United States; [23]Columbia University Vagelos College of Physicians and Surgeons, New York, NY, United States; [24]Erasmus University Medical Center, Rotterdam, Netherlands; [25]BridgeBio Pharma Inc, San Francisco, CA, United States; [26]Massachusetts General Hospital, Boston, MA, United States; [27]Harvard Medical School, Boston, MA, United States

BACKGROUND

ADH1 is caused by *CASR* gain-of-function variants increasing calcium sensitivity in the parathyroid and kidney[1,2]

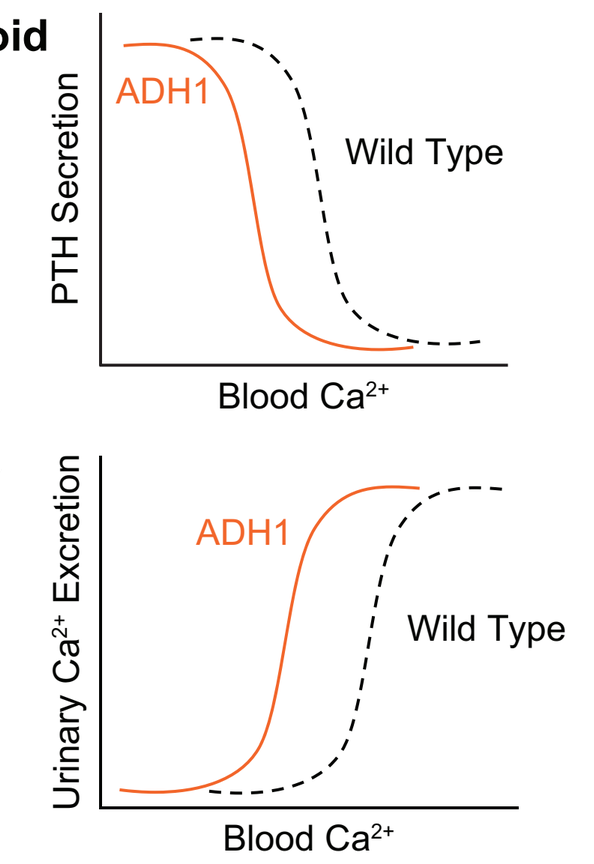
Parathyroid
ADH1
Wild Type
PTH Secretion
Blood Ca2+
Kidney
ADH1
Wild Type
Urinary Ca2+ Excretion
Blood Ca2+

Untreated ADH1 / conventional therapy	Goals of targeted therapy
Oversensitive CaSR	CaSR signaling normalized
Parathyroid: Low PTH secretion **Kidney:** Reduced tubular Ca^{2+} reabsorption	**Parathyroid:** PTH secretion restored **Kidney:** Tubular Ca^{2+} reabsorption increased
Chronic hypocalcemia and persistent hypercalciuria	Normalization of serum and urinary calcium
Hypocalcemia symptoms (eg, tetany, seizures, brain fog) and risk of nephrocalcinosis, nephrolithiasis, and impaired kidney function	Improved hypocalcemia symptoms and reduced long-term renal complications

Conventional therapy (calcium supplements and active vitamin D) does not correct the underlying pathophysiology and may leave patients exposed to hypocalcemia and renal complications

STUDY DESIGN AND PARTICIPANTS

Study Design: Phase 3 Study (CALIBRATE) – 25 sites, 9 countries

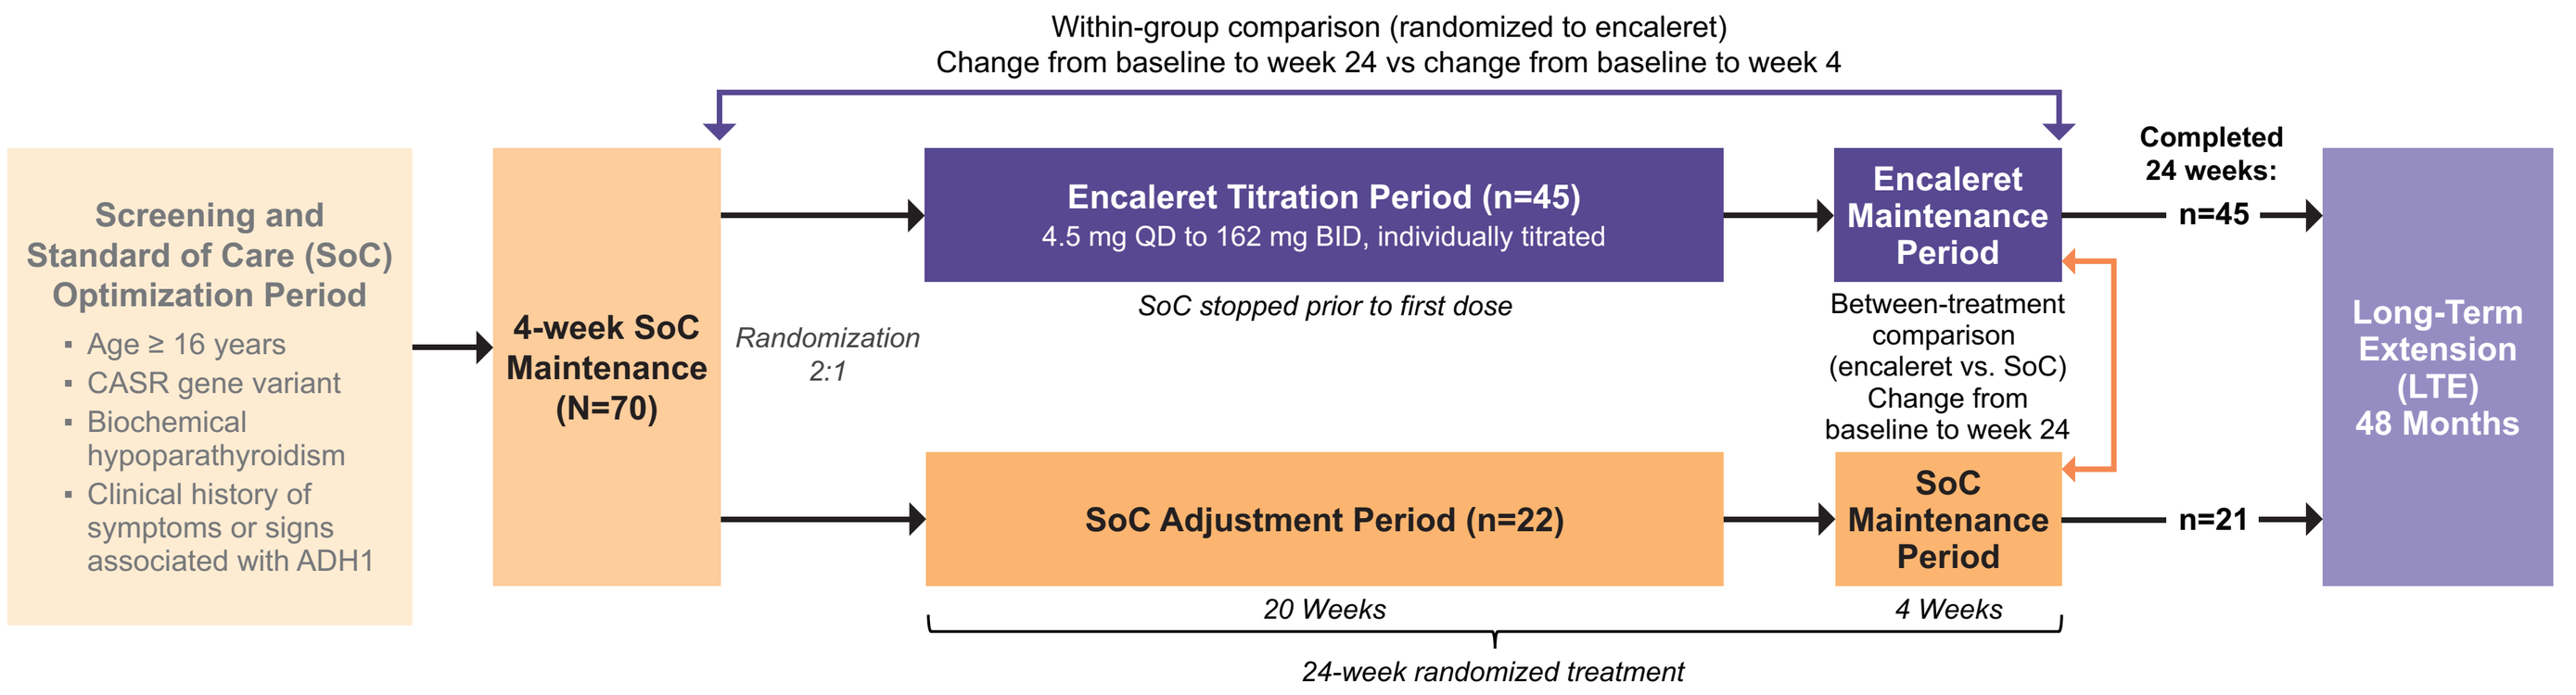
Within-group comparison (randomized to encaleret)
Change from baseline to week 24 vs change from baseline to week 4
Screening and Standard of Care (SoC) Optimization Period
Age ≥ 16 years
CASR gene variant
Biochemical hypoparathyroidism
Clinical history of symptoms or signs associated with ADH1
4-week SoC Maintenance (N=70)
Randomization 2:1
Encaleret Titration Period (n=45)
4.5 mg QD to 162 mg BID, individually titrated
SoC stopped prior to first dose
Encaleret Maintenance Period
Between-treatment comparison (encaleret vs. SoC) Change from baseline to week 24
SoC Adjustment Period (n=22)
SoC Maintenance Period
20 Weeks
4 Weeks
24-week randomized treatment
Completed 24 weeks:
n=45
n=21
Long-Term Extension (LTE) 48 Months

Primary Composite Endpoint at 24 weeks (within encaleret-treated group):
- Proportion of participants achieving:
 - cCa 8.3-10.7 mg/dL
 AND
 - UCa <300 mg/day (male)/<250 mg/day (female)

Additional Secondary and Exploratory Endpoints:
- cCa and 24-hour UCa between-treatment groups
- iPTH ≥ 15 pg/mL
- Phosphorus and magnesium
- **eGFR**
- **Nephrocalcinosis and nephrolithiasis assessed via renal ultrasound**

SoC = standard of care; a combination of oral active Vitamin D and/or calcium supplements. *CASR* = calcium-sensing receptor gene. iPTH = intact parathyroid hormone. eGFR = estimated glomerular filtration rate. cCa = albumin-corrected serum calcium; UCa = 24-hour urinary calcium. 99 participants were screened, 70 enrolled, and 67 randomized (June 2023–March 2025). Ultrasounds performed locally on prequalified equipment and read by a central reader blinded to study treatment and timepoint.

- **Current analysis:** Evaluation of the effect of encaleret, an investigational oral calcilytic, on renal endpoints — 24-hour urinary Ca^{2+}, eGFR, and renal ultrasound findings

Baseline characteristics

Characteristic	Target / Reference Range	Encaleret (N=45)	SoC (N=22)
Age (years)	—	43.7 ± 16.0	36.8 ± 13.4
Female sex, n (%)	—	24 (53)	11 (50)
***CASR* variants, distinct*, n**	—	34	17
Mineral parameters			
cCa (mg/dL)	8.3–10.7	8.4 ± 0.8	7.9 ± 0.7
24-hour uCa (mg/day)	<300 men, <250 women	397 ± 178	331 ± 195
iPTH (pg/mL)	15–65	6.8 ± 7.8	6.0 ± 4.5
Phosphorus (mg/dL)	2.5–4.8	4.8 ± 0.7	5.0 ± 1.0
Magnesium (mg/dL)	1.8–2.4	1.8 ± 0.2	1.7 ± 0.1
Kidney function and imaging			
eGFR (CKD-EPI)** (mL/min/1.73 m^2)	≥ 90	77 ± 23 (n=42)	83 ± 27 (n=21)
eGFR (bedside Schwartz)** (mL/min/1.73 m^2)	≥ 90	82 ± 23 (n=3)	97 (n=1)
Nephrocalcinosis***, n (%)	—	35 (80)	17 (81)
Nephrolithiasis***, n (%)	—	0 (0)	1 (4.8)
History of nephrolithiasis, n (%)	—	10 (22)	4 (18)
Conventional therapy****			
Elemental calcium (mg/day)	—	1953 ± 2379 (n=40)	1912 ± 2016 (n=20)
Calcitriol (µg/day)	—	0.90 ± 0.45 (n=36)	0.97 ± 0.86 (n=17)
Alfacalcidol (µg/day)	—	2.82 ± 1.03 (n=11)	2.06 ± 1.74 (n=4)

Data represent mean ± SD unless otherwise indicated. *Counts are of distinct *CASR* variants, not participants; 43 distinct variants were identified across the study population, of which 8 were represented in both treatment groups. **Determined via CKD-EPI for participants ≥18 years and via bedside Schwartz for the 4 participants <18 years. ***Assessed by renal ultrasound, performed at baseline in 65 participants (encaleret n=44, SoC n=21); percentages are based on participants with imaging available. Presence of nephrocalcinosis defined as grade 1 or higher. ****Mean total daily doses averaged over Period 1.

TREATMENT OUTCOMES

76% of participants met the primary endpoint after 24 weeks of encaleret

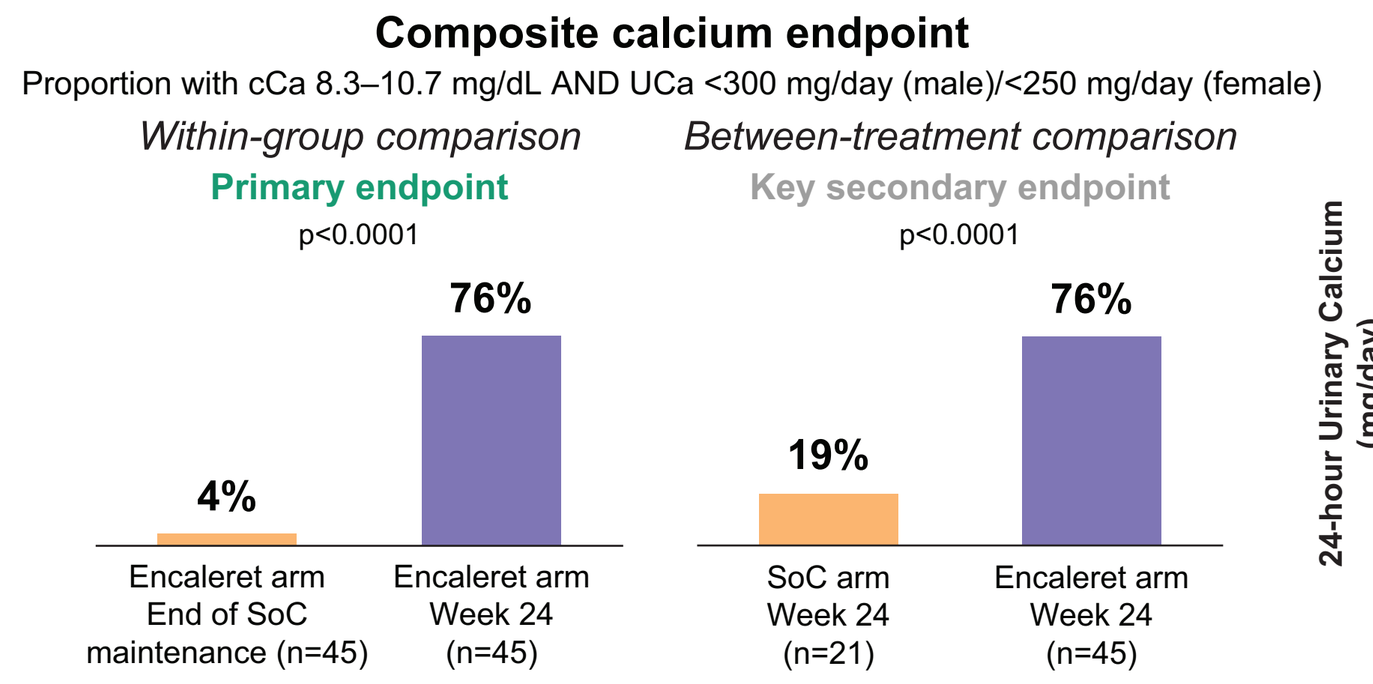
Composite calcium endpoint
Proportion with cCa 8.3–10.7 mg/dL AND UCa <300 mg/day (male)/<250 mg/day (female)
Within-group comparison
Primary endpoint
p<0.0001
Between-treatment comparison
Key secondary endpoint
p<0.0001
4%
76%
19%
76%
Encaleret arm End of SoC maintenance (n=45)
Encaleret arm Week 24 (n=45)
SoC arm Week 24 (n=21)
Encaleret arm Week 24 (n=45)

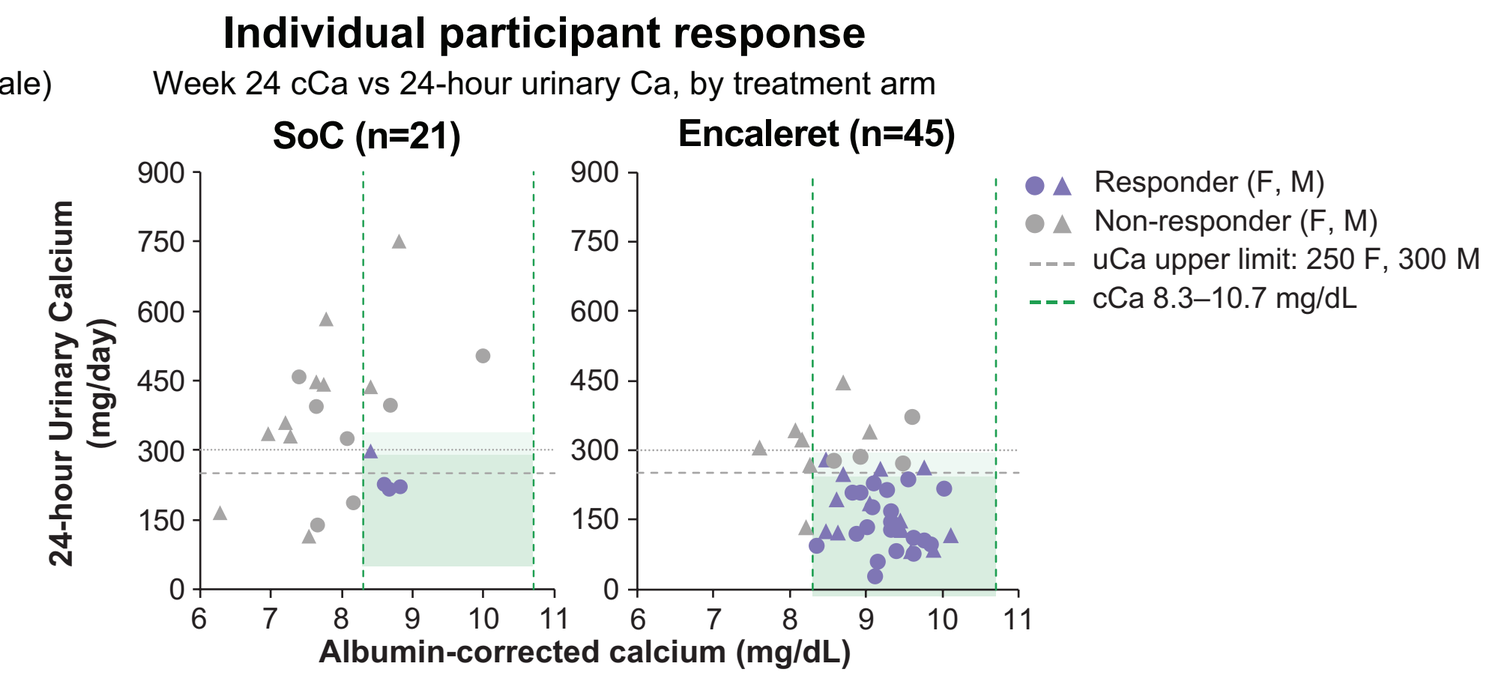
Individual participant response
Week 24 cCa vs 24-hour urinary Ca, by treatment arm
SoC (n=21)
Encaleret (n=45)
24-hour Urinary Calcium (mg/day)
Albumin-corrected calcium (mg/dL)
Responder (F, M)
Non-responder (F, M)
uCa upper limit: 250 F, 300 M
cCa 8.3–10.7 mg/dL

Encaleret restored PTH secretion, with changes maintained over 24 weeks

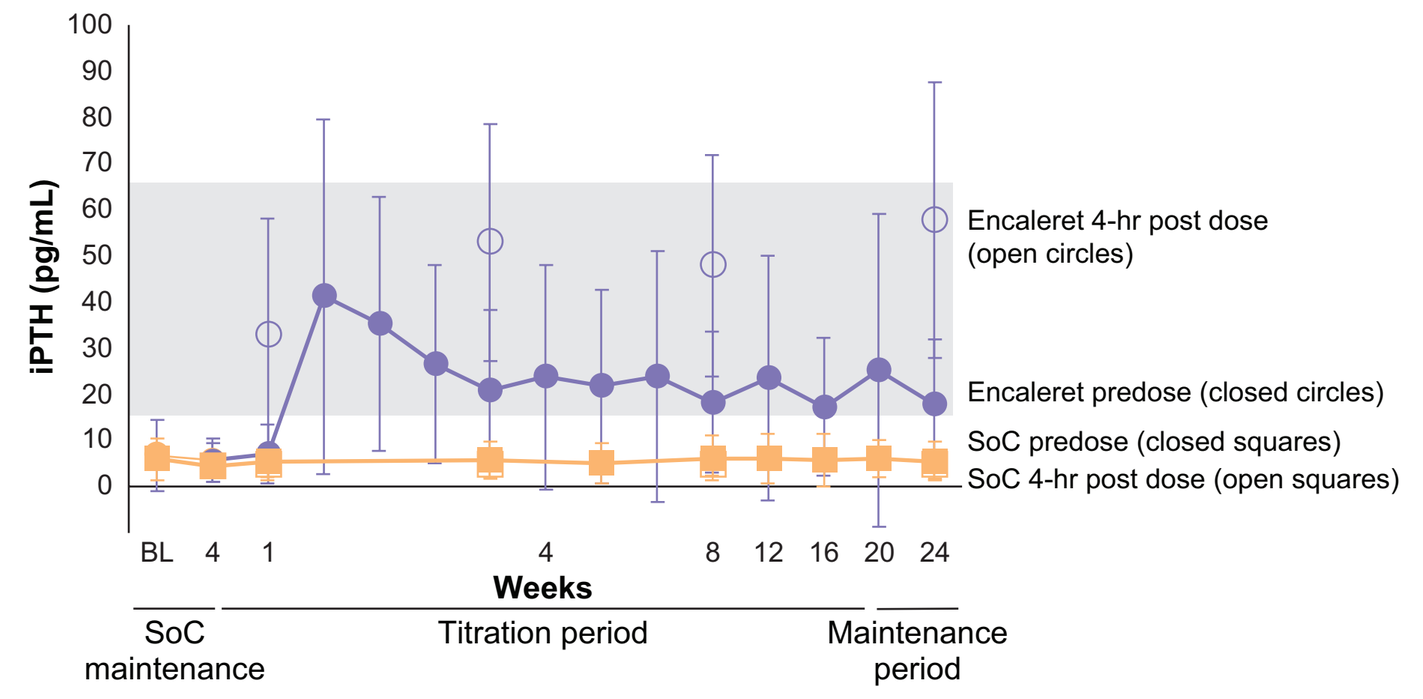
iPTH (pg/mL)
Encaleret 4-hr post dose (open circles)
Encaleret predose (closed circles)
SoC predose (closed squares)
SoC 4-hr post dose (open squares)
Weeks
SoC maintenance
Titration period
Maintenance period

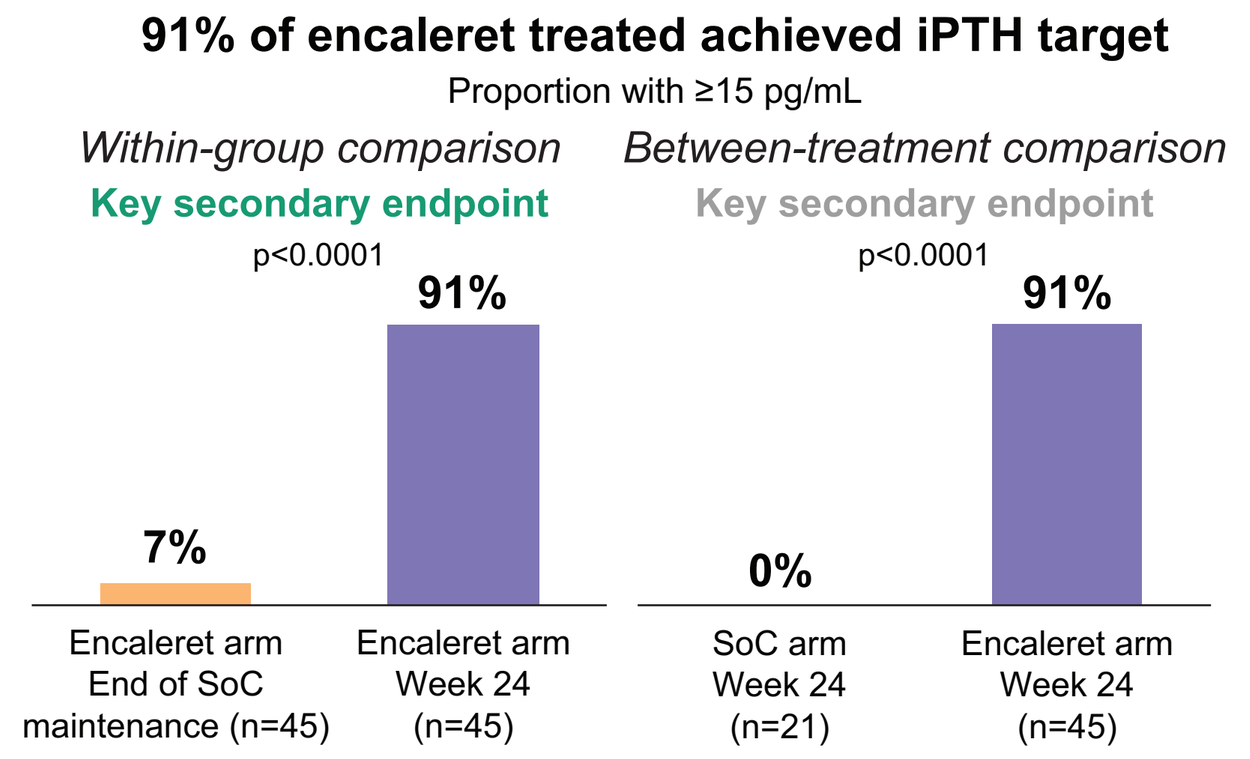
91% of encaleret treated achieved iPTH target
Proportion with ≥15 pg/mL
Within-group comparison
Key secondary endpoint
p<0.0001
Between-treatment comparison
Key secondary endpoint
p<0.0001
7%
91%
0%
91%
Encaleret arm End of SoC maintenance (n=45)
Encaleret arm Week 24 (n=45)
SoC arm Week 24 (n=21)
Encaleret arm Week 24 (n=45)

iPTH Week 24 ΔBL: Encaleret 50.8 ± 29.1 vs SoC -1.3 ± 3.1 pg/mL
Between-group difference 52.2 (SE 4.7), p<0.0001
Encaleret within-arm: ΔBL to W24 vs ΔBL to W4 on SoC maintenance, p<0.0001

Data reported as mean ± SD. Gray shading denotes the target or reference range.

Encaleret increased serum calcium, with changes maintained over 24 weeks

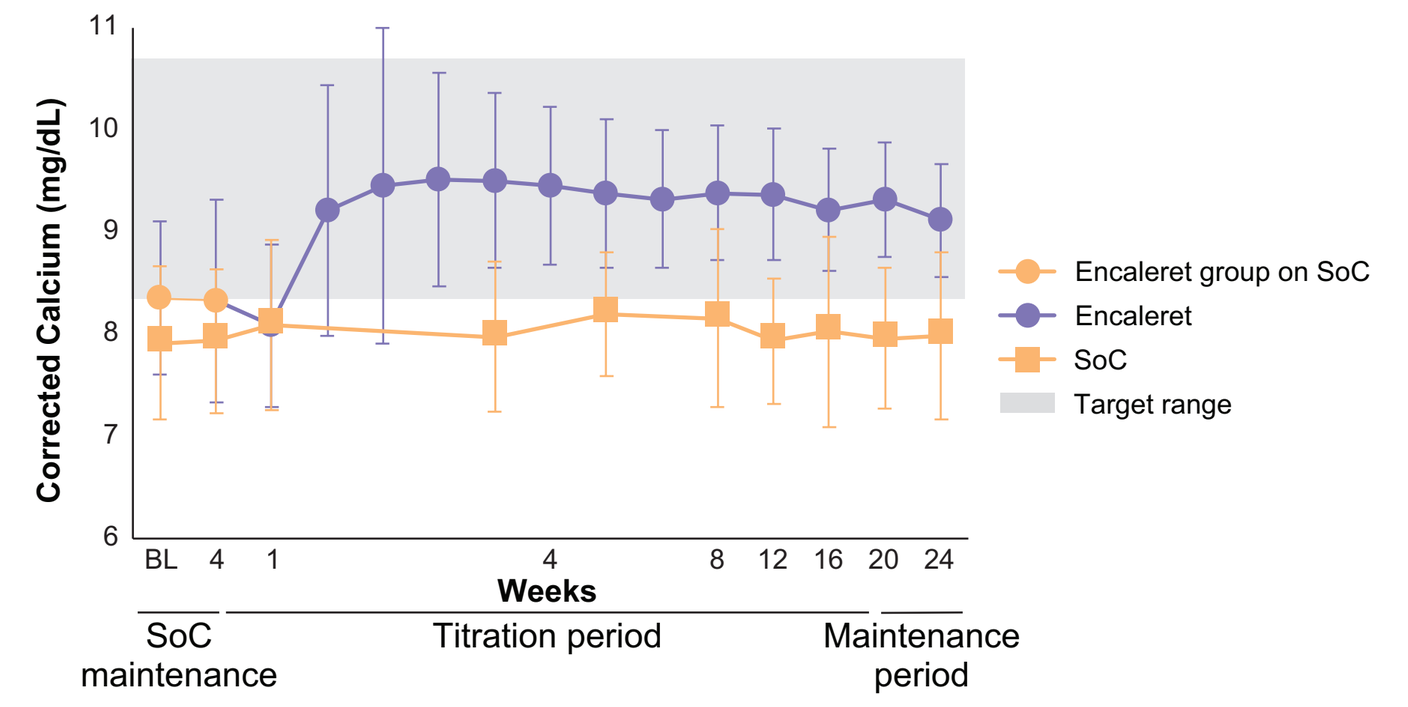
Corrected Calcium (mg/dL)
Encaleret group on SoC
Encaleret
SoC
Target range
Weeks
SoC maintenance
Titration period
Maintenance period

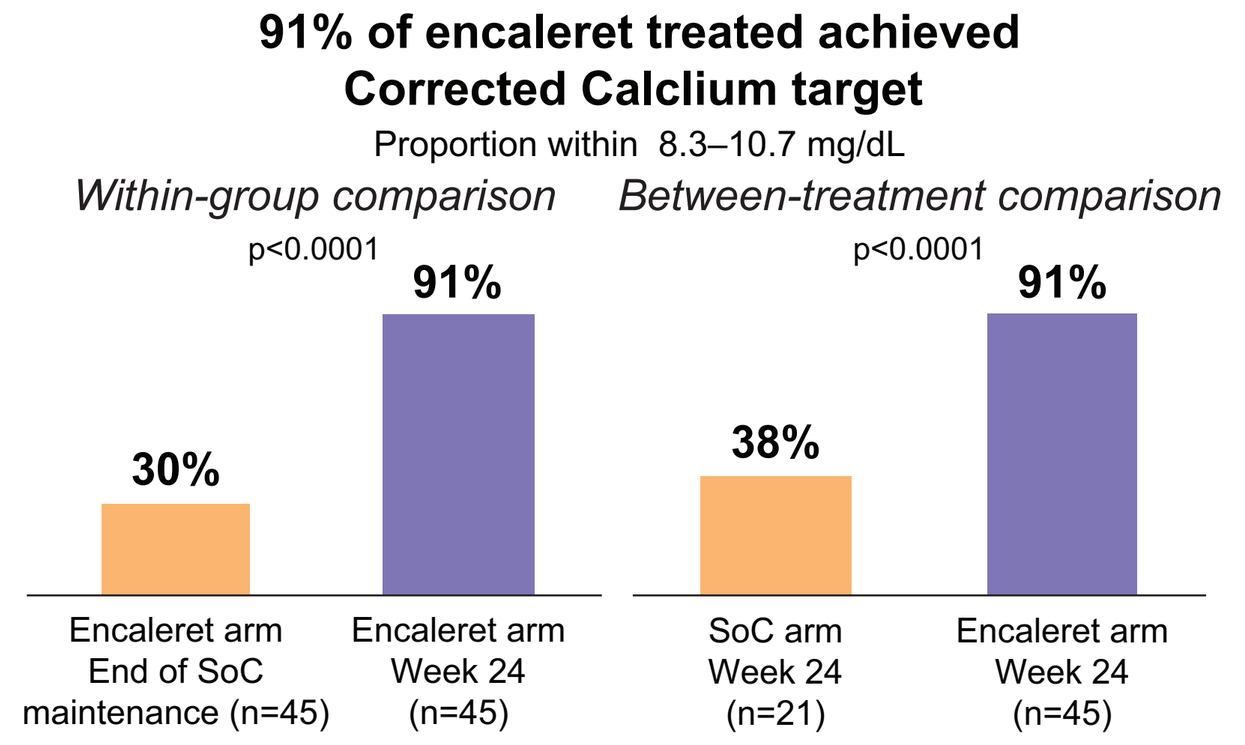
91% of encaleret treated achieved Corrected Calclium target
Proportion within 8.3–10.7 mg/dL
Within-group comparison
p<0.0001
Between-treatment comparison
p<0.0001
30%
91%
38%
91%
Encaleret arm End of SoC maintenance (n=45)
Encaleret arm Week 24 (n=45)
SoC arm Week 24 (n=21)
Encaleret arm Week 24 (n=45)

cCa Week 24 ΔBL: Encaleret +0.7 ± 0.9 vs SoC +0.1 ± 0.9 mg/dL
Between-group difference 0.6 (SE 0.2), p<0.01
Encaleret within-arm: ΔBL to W24 vs ΔBL to W4 on SoC maintenance, p<0.0001

Data reported as mean ± SD. Gray shading denotes the target or reference range.

Encaleret decreased 24-hour urinary calcium over the study period

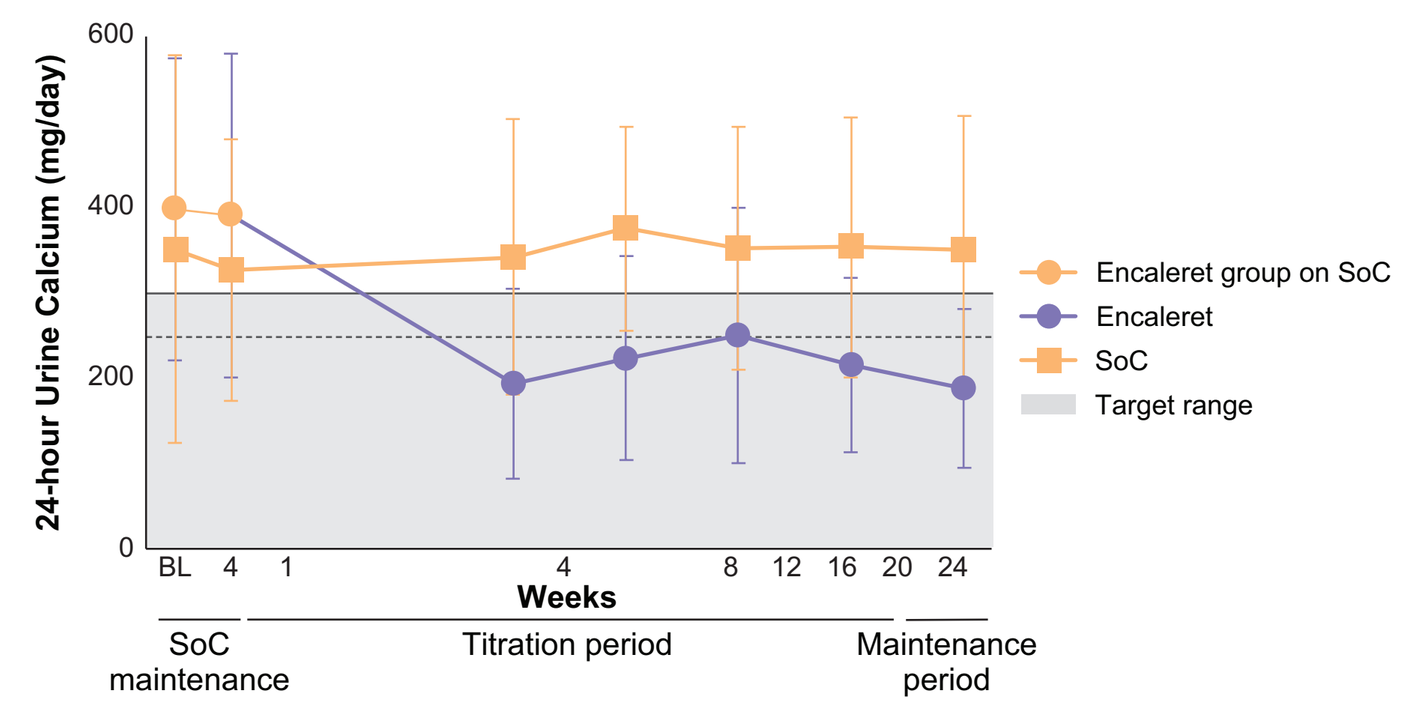
24-hour Urine Calcium (mg/day)
Encaleret group on SoC
Encaleret
SoC
Target range
Weeks
SoC maintenance
Titration period
Maintenance period

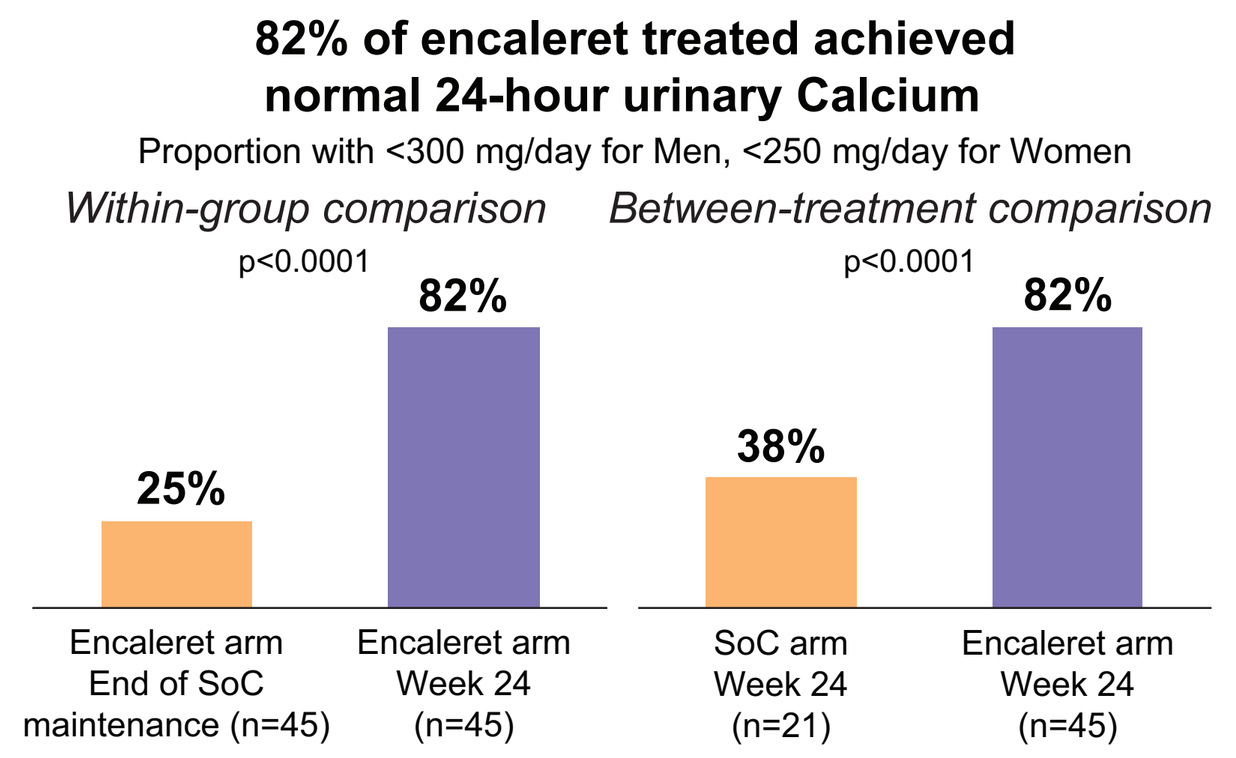
82% of encaleret treated achieved normal 24-hour urinary Calcium
Proportion with <300 mg/day for Men, <250 mg/day for Women
Within-group comparison
p<0.0001
Between-treatment comparison
p<0.0001
25%
82%
38%
82%
Encaleret arm End of SoC maintenance (n=45)
Encaleret arm Week 24 (n=45)
SoC arm Week 24 (n=21)
Encaleret arm Week 24 (n=45)

24-hour uCa ΔBL at Week 24: Encaleret –207 ± 174 vs SoC +11 ± 186 mg/day
Between-group difference –221 (SE 47), p<0.0001
Encaleret within-arm: ΔBL to W24 vs ΔBL to W4 on SoC maintenance, p<0.0001

For 24-hour urine calcium, the solid line reflects the upper limit for men and the dashed line the upper limit for women.

Encaleret reduced serum phosphorus and increased serum magnesium over 24 weeks

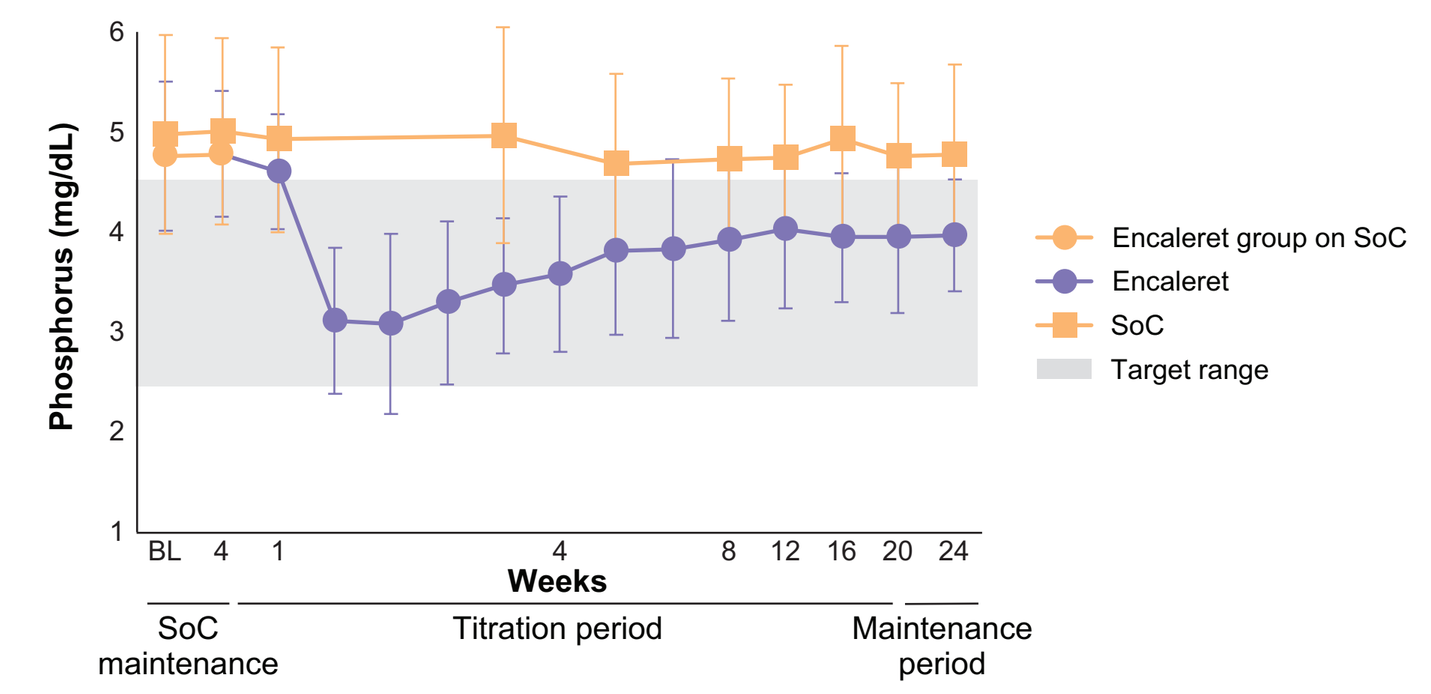
Phosphorus (mg/dL)
Encaleret group on SoC
Encaleret
SoC
Target range
Weeks
SoC maintenance
Titration period
Maintenance period

Phosphorus Week 24 ΔBL: Encaleret –0.8 ± 0.7 vs SoC –0.2 ± 0.8 mg/dL
Between-group difference –0.6 (SE 0.2), p<0.01
Encaleret within-arm: ΔBL to W24 vs ΔBL to W4 on SoC maintenance, p<0.0001

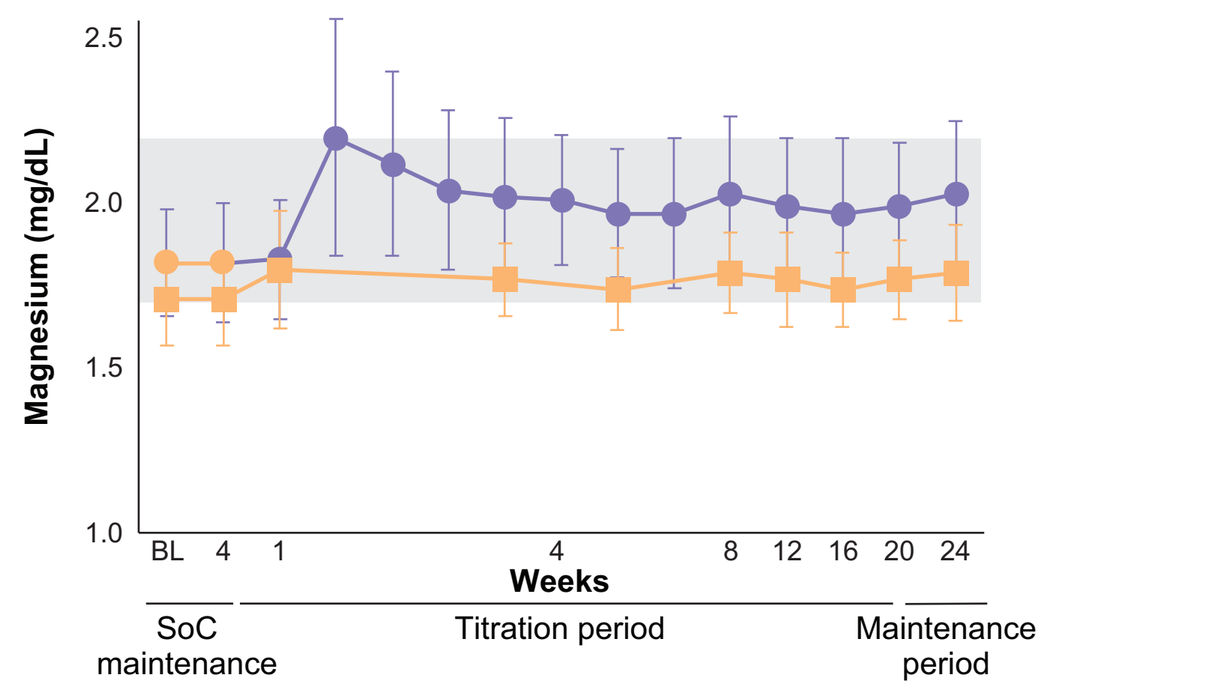
Magnesium (mg/dL)
Weeks
SoC maintenance
Titration period
Maintenance period

Magnesium Week 24 ΔBL: Encaleret +0.2 ± 0.2 vs SoC +0.1 ± 0.1 mg/dL
Between-group difference 0.13 (SE 0.04), p<0.01
Encaleret within-arm: ΔBL to W24 vs ΔBL to W4 on SoC maintenance, p<0.0001

Data reported as mean ± SD. Gray shading denotes the reference range.

Encaleret increased eGFR over the study period

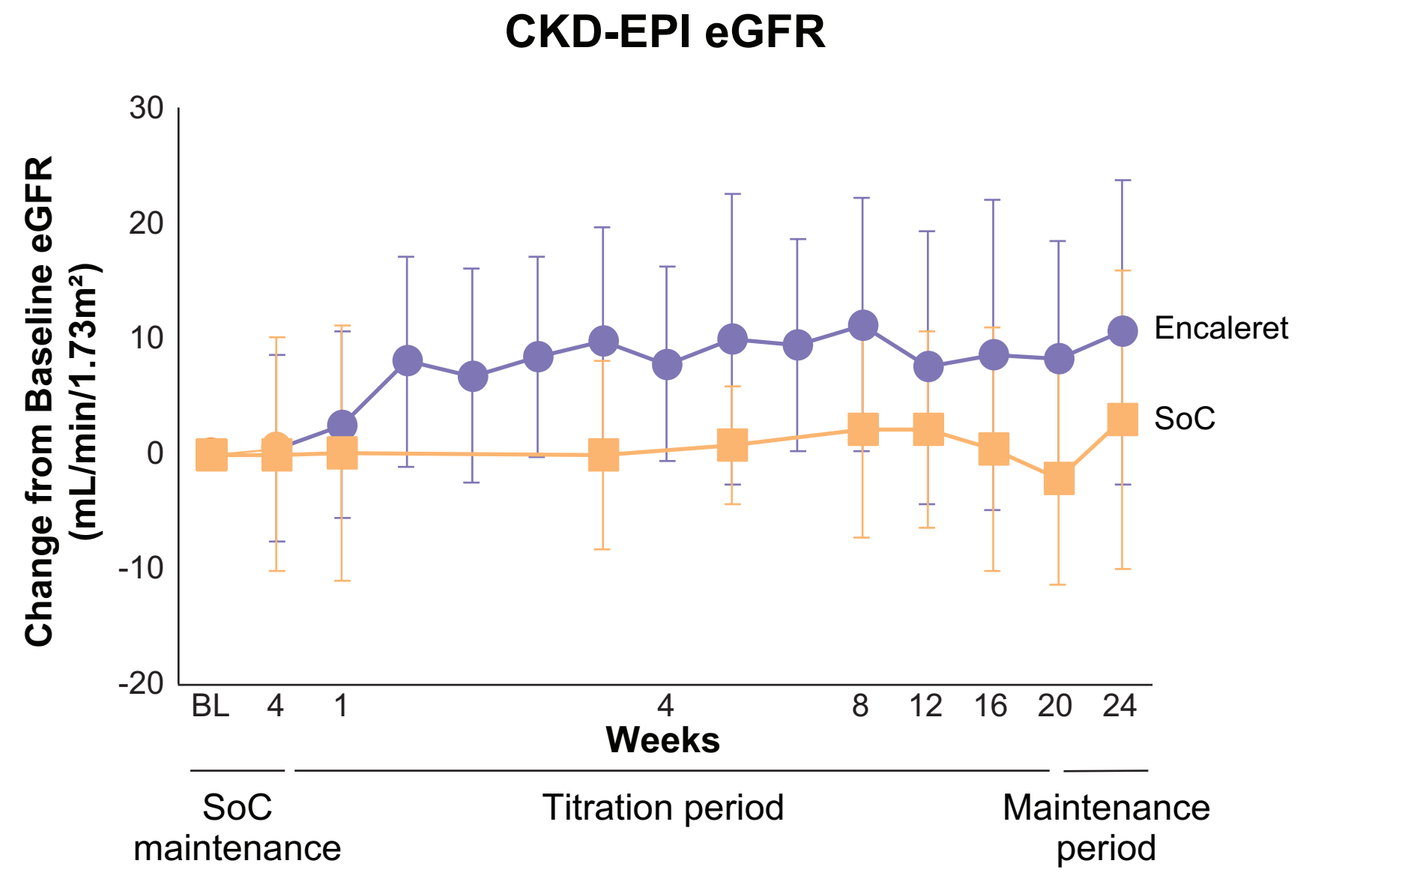
CKD-EPI eGFR
Change from Baseline eGFR (mL/min/1.73m²)
Encaleret
SoC
Weeks
SoC maintenance
Titration period
Maintenance period

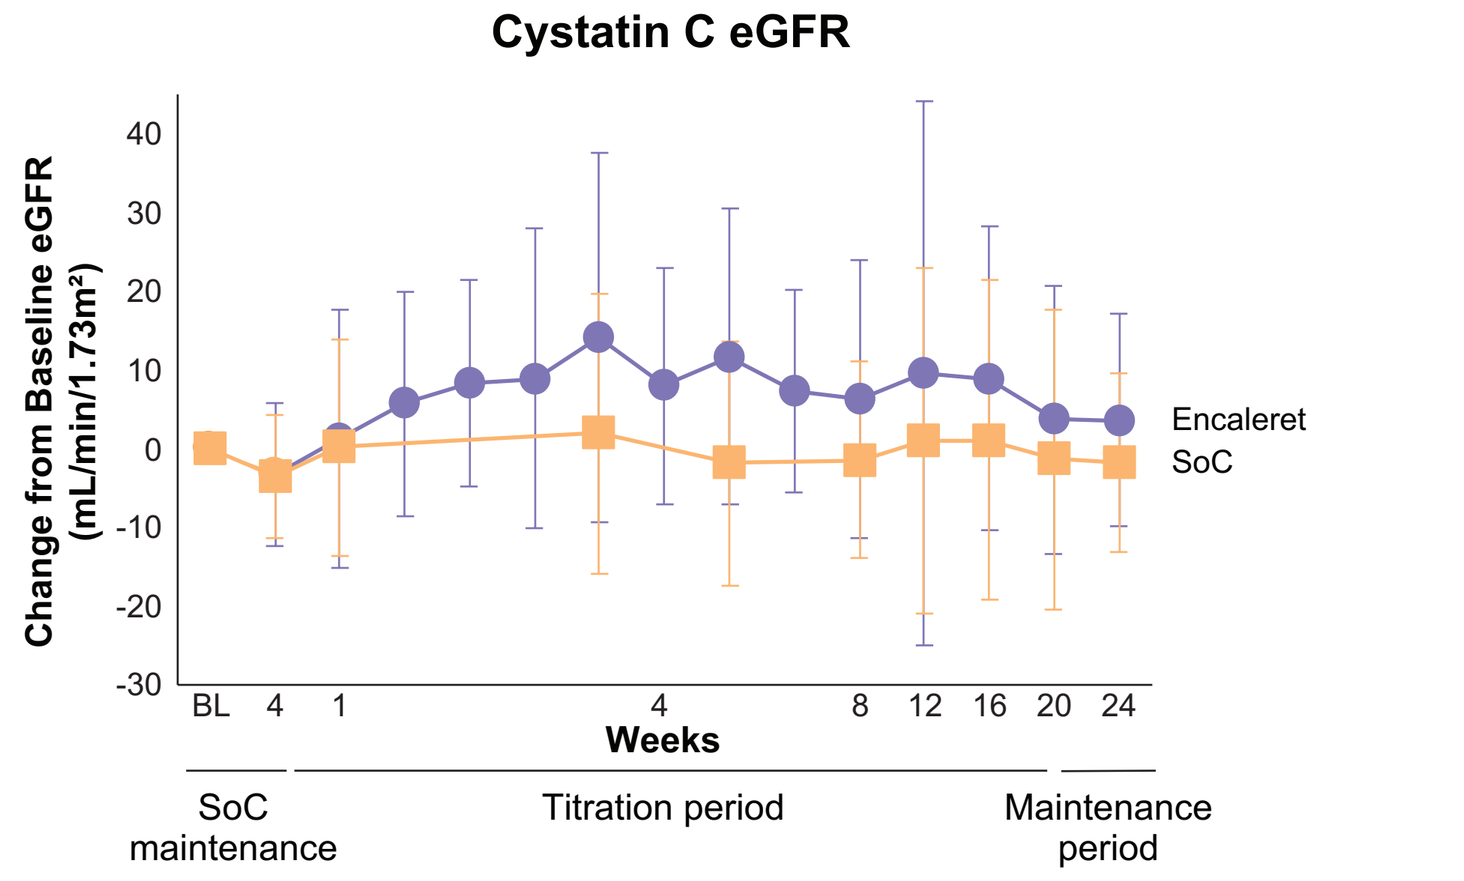
Cystatin C eGFR
Change from Baseline eGFR (mL/min/1.73m²)
Encaleret
SoC
Weeks
SoC maintenance
Titration period
Maintenance period

Week 24 ΔBL: Encaleret +11±13 vs SoC +3±13 mL/min/1.73m²
Between-group difference 7.6 (SE 3.6), p<0.05
Encaleret within-arm: ΔBL to W24 vs ΔBL to W4 on SoC maintenance, p<0.0001

Line graphs represent mean ± SD throughout the study for change from baseline in eGFR calculated by CKD-EPI; data from four participants <18 years whose eGFR was calculated via bedside Schwartz were excluded. Samples were collected following an 8-hour fast and prior to the morning encaleret or SoC dose.

eGFR CKD stage improved or remained stable in 98% of encaleret participants vs 85% with SoC

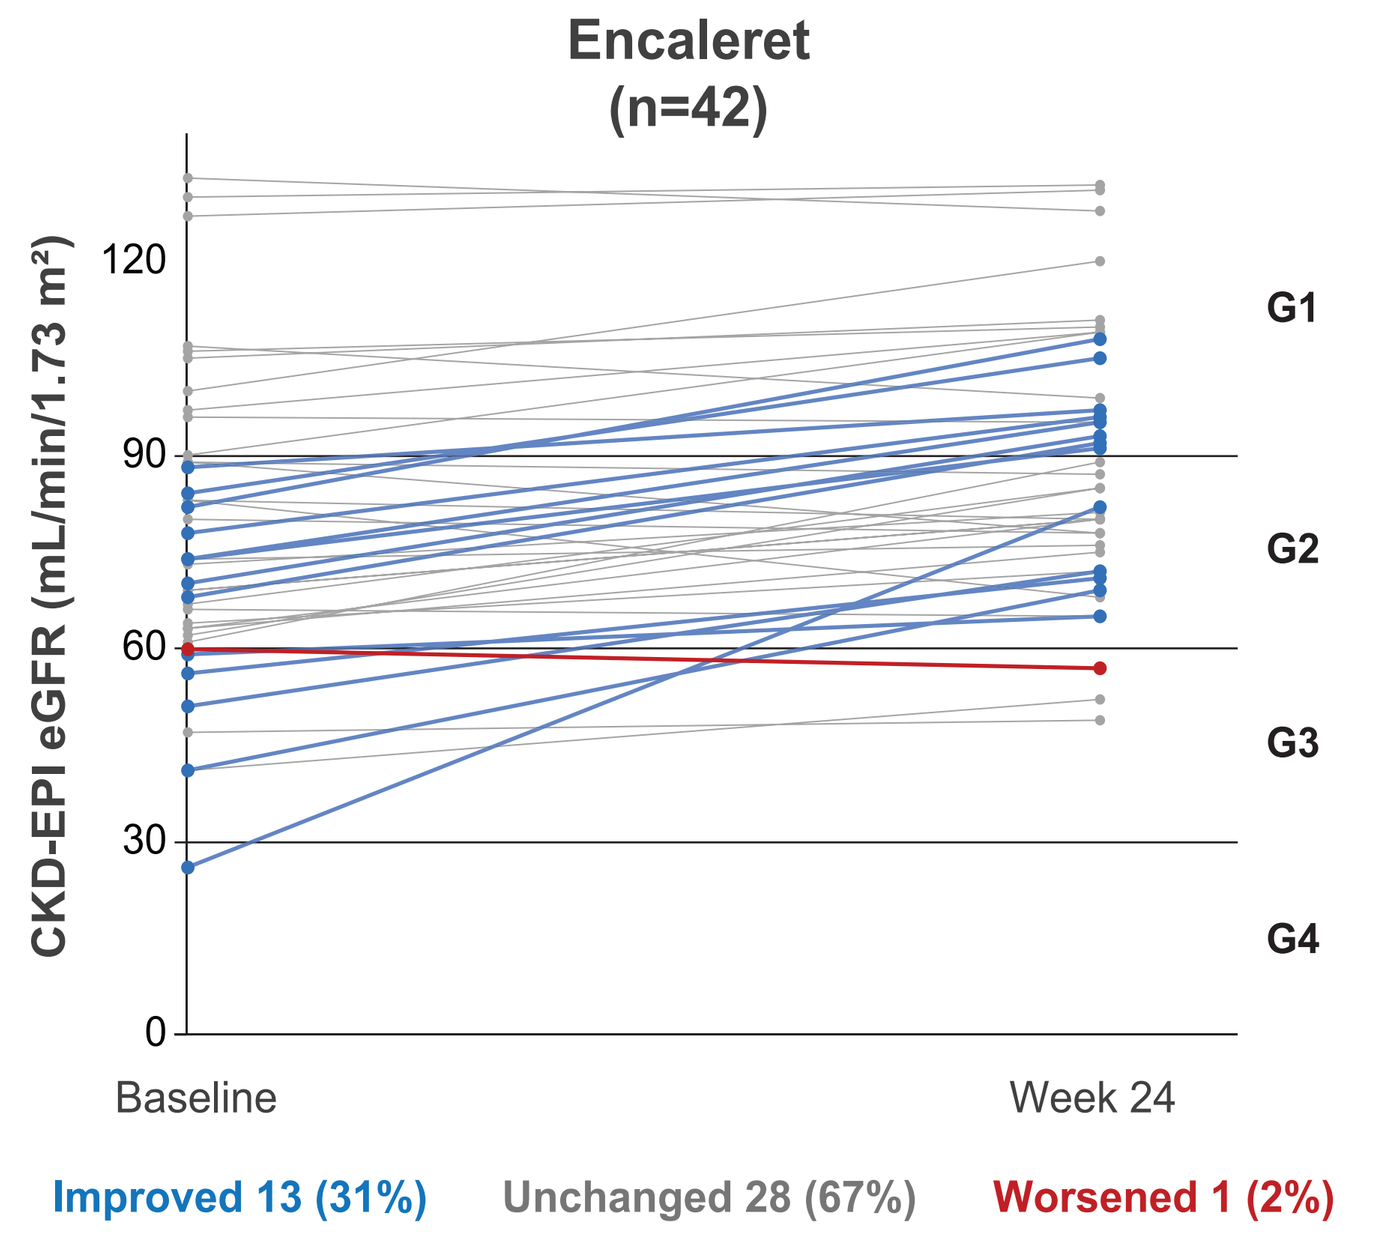
Encaleret (n=42)
CKD-EPI eGFR (mL/min/1.73 m²)
G1
G2
G3
G4
Baseline
Week 24
Improved 13 (31%)
Unchanged 28 (67%)
Worsened 1 (2%)

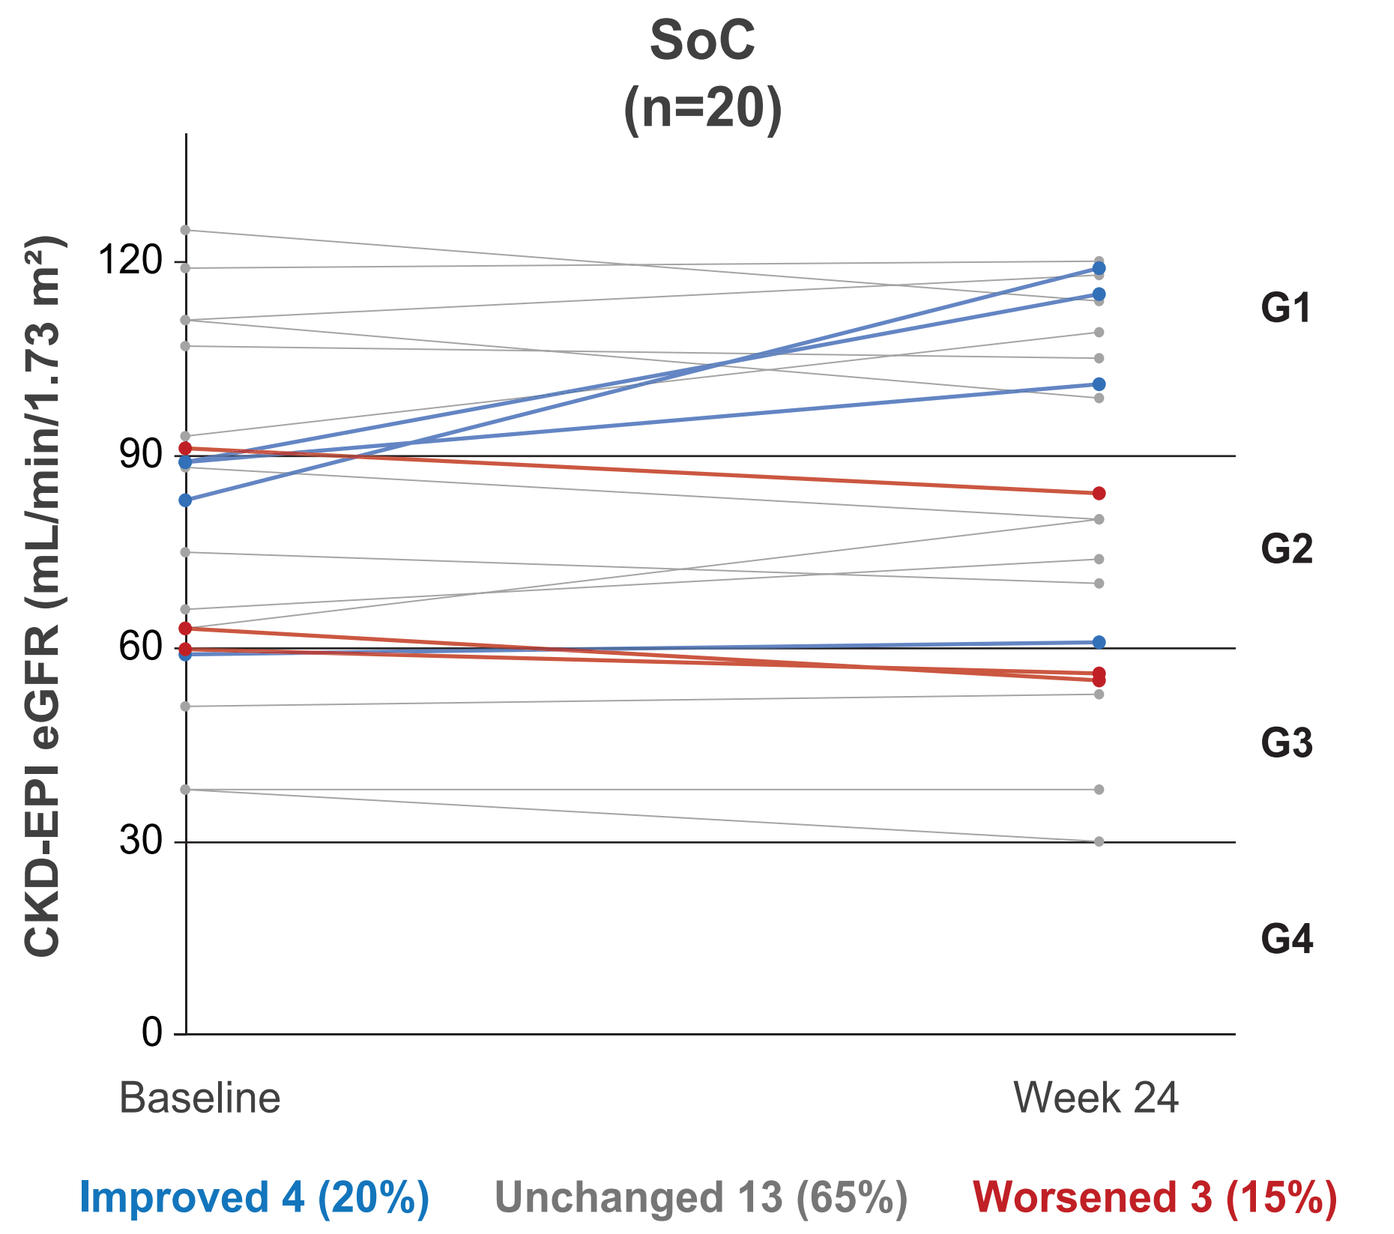
SoC (n=20)
CKD-EPI eGFR (mL/min/1.73 m²)
G1
G2
G3
G4
Baseline
Week 24
Improved 4 (20%)
Unchanged 13 (65%)
Worsened 3 (15%)

CKD stage boundaries at eGFR 90, 60 and 30 mL/min/1.73 m²; line color shows direction of CKD stage change from baseline to Week 24. Participants <18 years (bedside Schwartz) and one SoC participant without a Week 24 value are excluded.

Nephrocalcinosis and nephrolithiasis assessed via renal ultrasound over 24 weeks

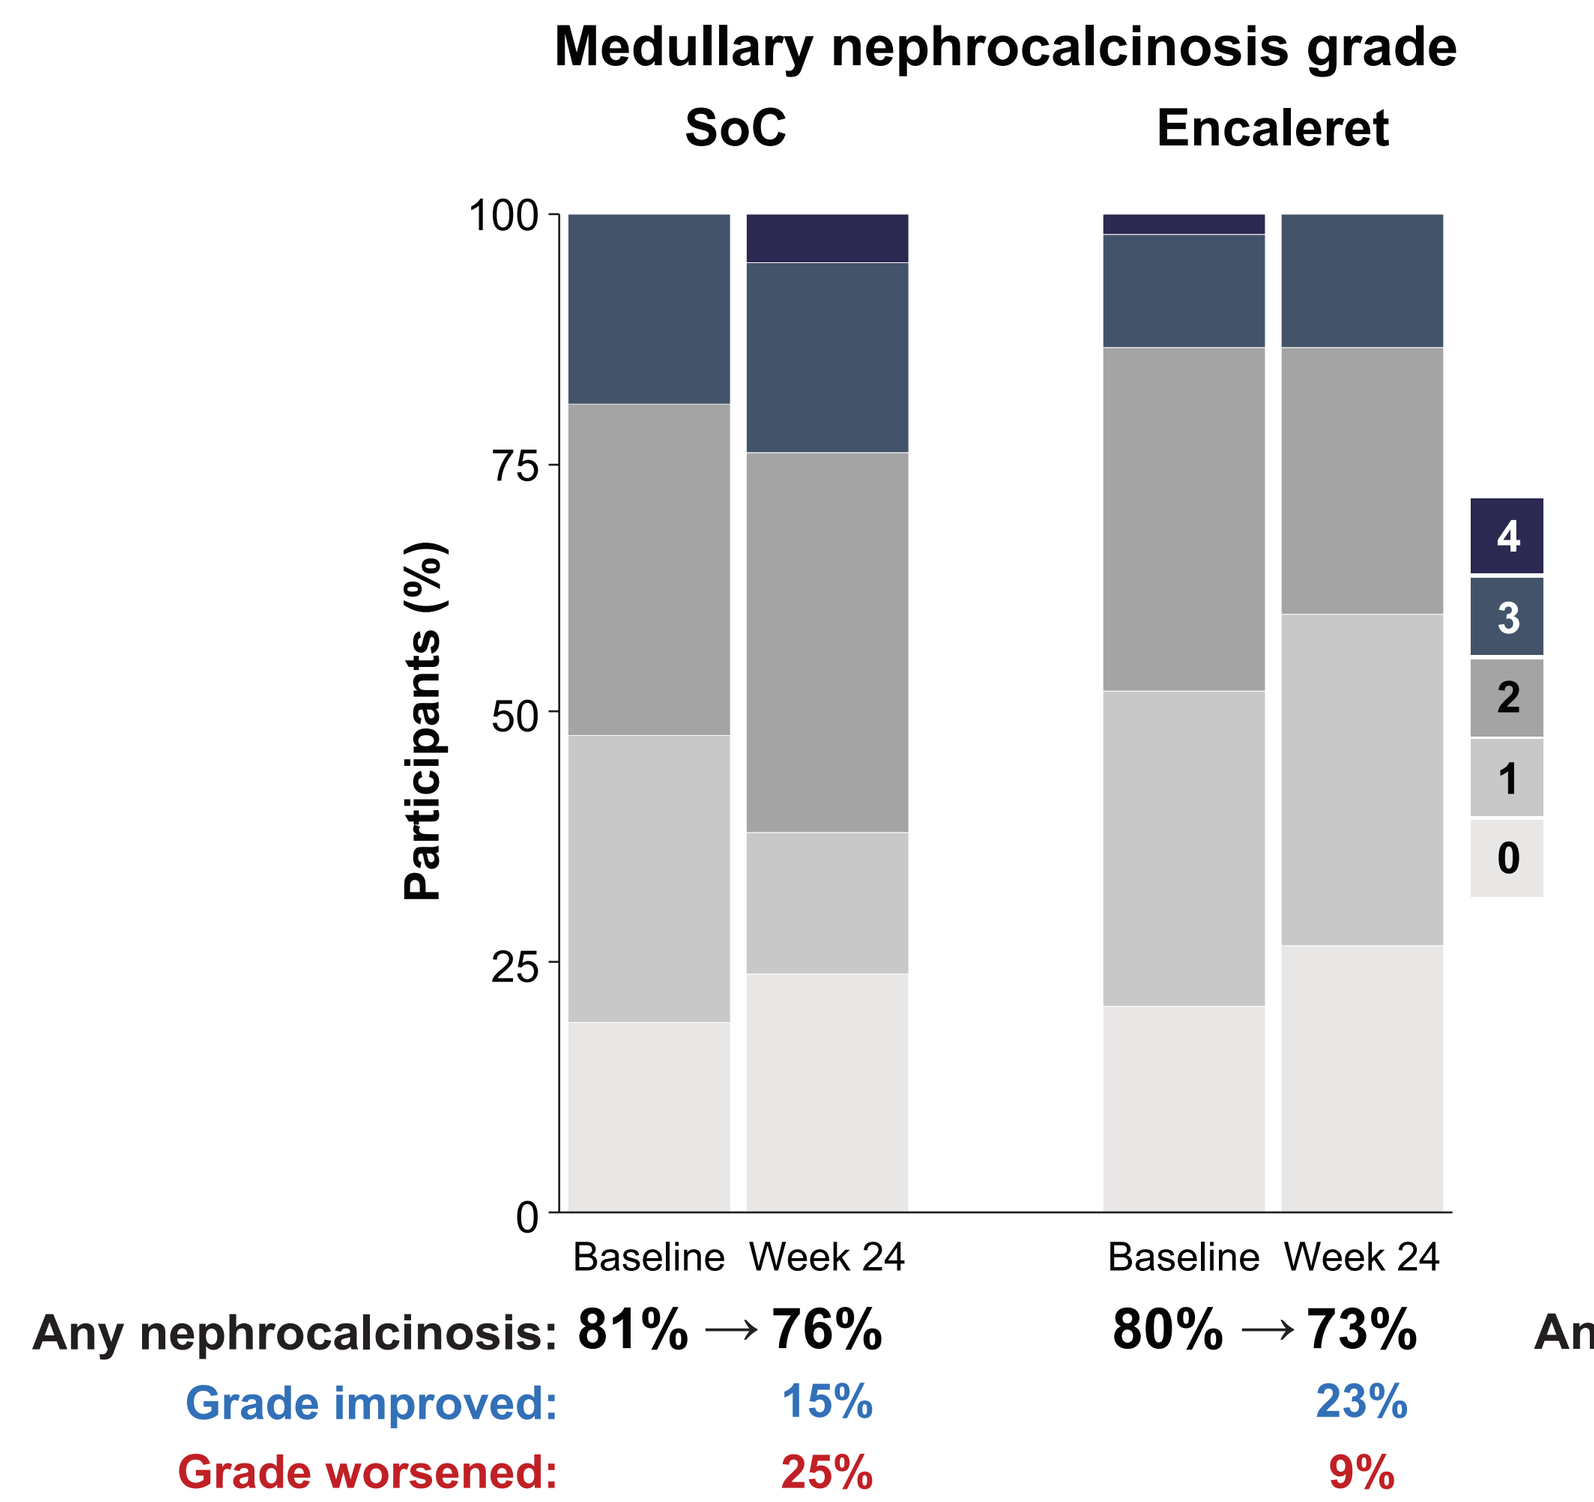
Medullary nephrocalcinosis grade
SoC
Encaleret
Participants (%)
Baseline
Week 24
4
3
2
1
0
Any nephrocalcinosis: 81% → 76%
80% → 73%
Grade improved: 15%
23%
Grade worsened: 25%
9%

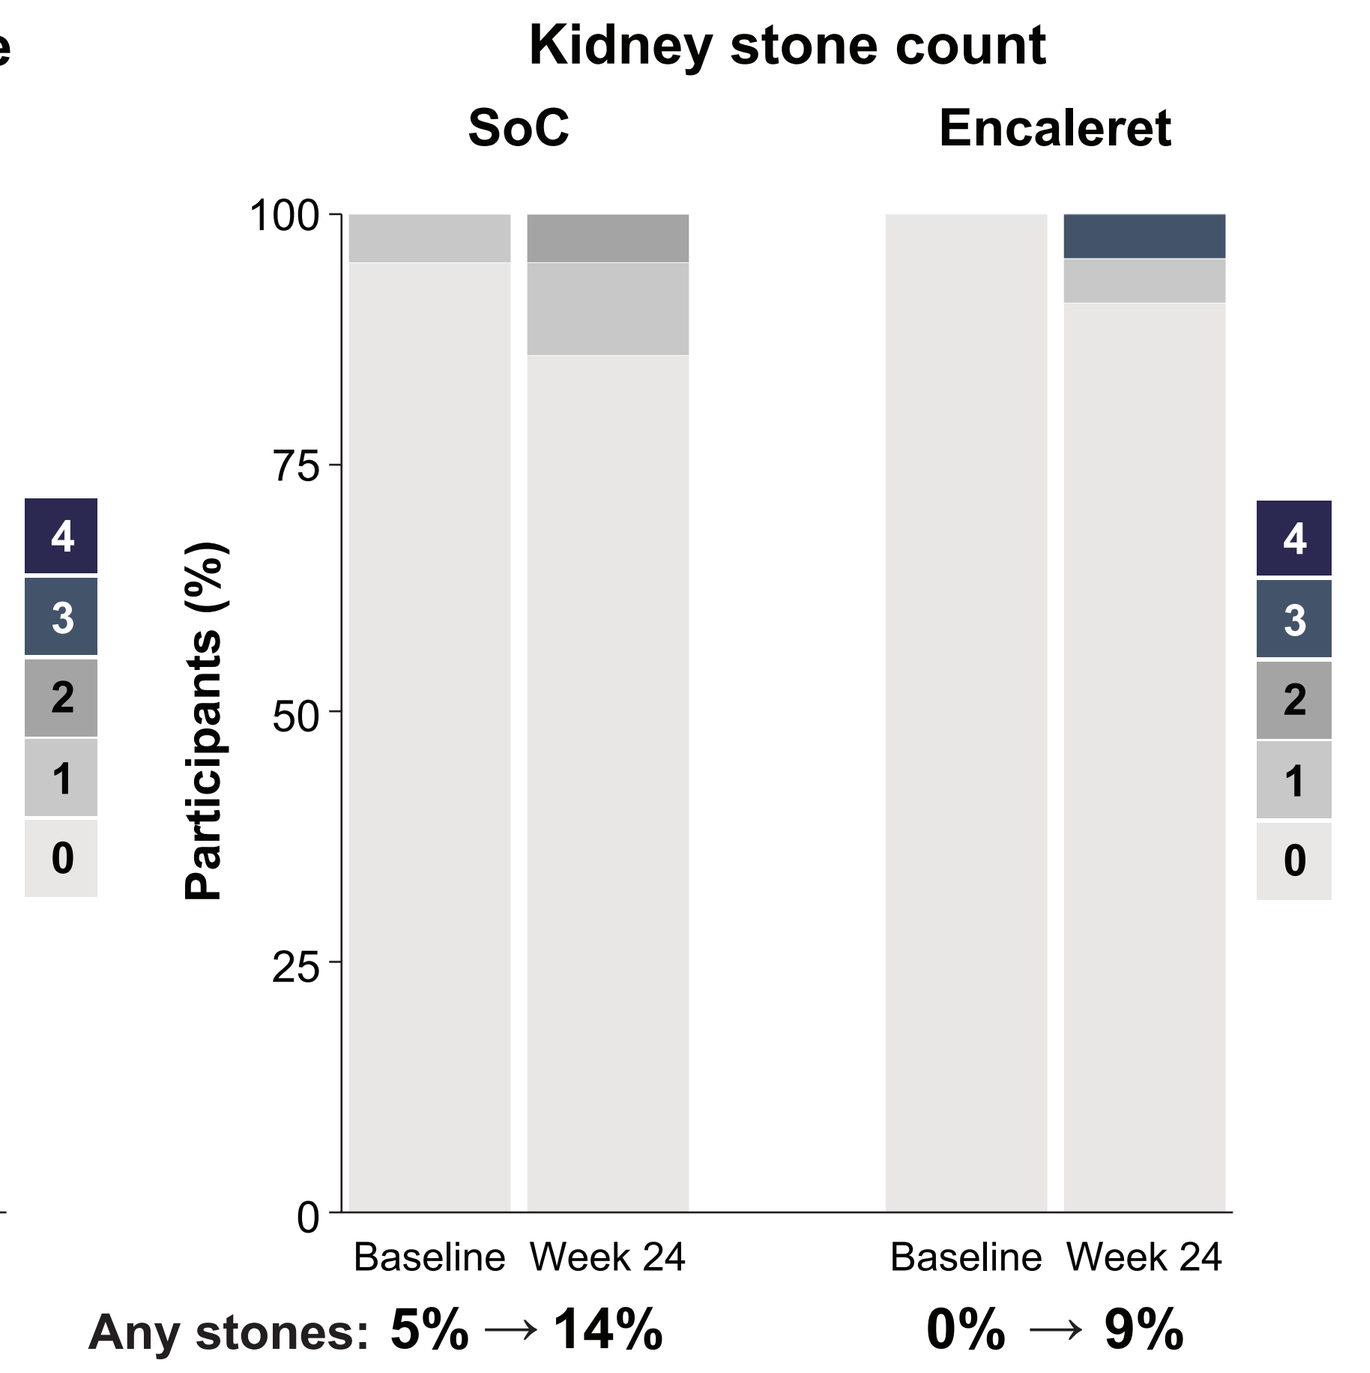
Kidney stone count
SoC
Encaleret
Participants (%)
Baseline
Week 24
4
3
2
1
0
Any stones: 5% → 14%
0% → 9%

Proportion of participants with each finding on renal ultrasound. Percentages are based on participants with imaging available (encaleret n=44 at baseline, n=45 at Week 24; SoC n=21). Stone count is the total across both kidneys; nephrocalcinosis grade is the highest grade in either kidney. Grade 4 was defined as diffuse calcifications with accumulation at the tips of nearly all renal pyramids. Nephrolithiasis was defined as a cortical stone or an isolated calcification within or at the tip of a pyramid when the remaining pyramids did not meet Grade 4 criteria. Imaging outcomes are descriptive and no between-group statistical comparison was performed.

- Overall nephrocalcinosis prevalence remained stable; kidney stone findings involved few participants, most of whom had history of nephrolithiasis
 - Nephrolithiasis: encaleret 0 to 4 participants; SoC 1 to 3 participants

No TEAEs led to encaleret discontinuation during randomized treatment

Overall safety

	SoC Maintenance (4 weeks)	Randomized treatment (24 weeks)	
	N = 67 n (%)	SoC N = 22 n (%)	Encaleret N = 45 n (%)
Any TEAE	30 (45)	14 (64)	40 (89)
Mild	23 (34)	6 (27)	21 (47)
Moderate	4 (6)	6 (27)	16 (36)
Severe	3 (5)	2 (9)	3 (7)
Serious TEAE	2 (3)	3 (14)	4 (9)
Related TEAE			
Any	4 (6)	0 (0)	16 (36)
Serious	1 (2)	0 (0)	1 (2)
Study Discontinuation	0 (0)	0 (0)	0 (0)
Hypercalcemia	3 (4.5)	0 (0)	10 (22.2)
Headache	0 (0)	1 (4.5)	9 (20.0)
Constipation	0 (0)	2 (9.1)	5 (11.1)
Nausea	1 (1.5)	3 (13.6)	4 (8.9)
Fatigue	0 (0)	1 (4.5)	4 (8.9)
Hypertension	0 (0)	1 (4.5)	4 (8.9)
Hypocalcemia*	4 (6.0)	3 (13.6)	3 (6.7)
Urinary tract infection	3 (4.5)	1 (4.5)	3 (6.7)
Sinusitis	0 (0)	2 (9.1)	2 (4.4)

TEAE = Treatment-Emergent Adverse Event. For each category, participants are included only once, even if they experienced multiple events in that category. Relatedness considered by the investigator as related to investigational product being administered in the respective study period reported. *Hypocalcemia was reported as TEAE if considered by investigator to have worsened from baseline. Data extracted on 15Oct2025.

CONCLUSIONS

- Encaleret normalized mean blood and urine calcium in most participants and was associated with an increase in mean eGFR in individuals with ADH1 at Week 24
 - Nephrocalcinosis was stable in both arms over 24 weeks
- Consistent with inhibition of the oversensitive renal CaSR, encaleret corrected hypocalcemia while also decreasing urinary calcium
- Follow-up is needed to assess encaleret's long-term impact on renal calcifcations and function
 - Encaleret has the potential to become the first personalized therapy for ADH1, directly targeting the underlying mechanism of the disease

REFERENCES: **1.** Roszko KL, et al. *Front Physiol.* 2016. **2.** Riccardi D, et al. *Nat Rev Nephrol.* 2016;12(7):414-425.

FUNDING: This study was sponsored by BridgeBio Pharma, Inc., San Francisco, CA, US.

ACKNOWLEDGEMENTS: The authors would like to thank all the patients and their families, the CALIBRATE investigators, study sites, and support staff who participated in this research.

PRESENTING AUTHOR DISCLOSURES: TBD

Presented at the American Society of Nephrology Kidney Week 2026; October 21–25, 2026, Denver, CO